Differential Expression and Sequence Variants of Sodium Taurocholate Cotransporting Polypeptide Do Not Affect Hepatitis Delta Virus Inhibition by Bulevirtide

SAT-405

Simin Xu[1], Thomas Aeschbacher[1], Savrina Manhas[1], Chunfeng Li[1], Ross Martin[1], Yang Liu[1], Evguenia Maiorova[1], Hongmei Mo[1], Roberto Mateo[1]

[1]Gilead Sciences, Inc., Foster City, CA, USA

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Conclusions

- Six NTCP SNPs were observed with MAFs >1% in 1 or more racial groups from public datasets
 - Four of 6 SNPs identified from public datasets were also found in the 50 PHH donors, suggesting the SNPs from PHH donors can represent NTCP prevalence in the public dataset
 - Two of 6 SNPs not identified in the 50 PHH donors were nonsynonymous and had a low prevalence (<2%) in all racial groups
- BLV remained active against HDV on PHHs expressing different levels of NTCP and NTCP variants observed in the 50 US donors
- BLV remained active against HDV on Huh7 cell lines expressing variant NTCP-I168T and different levels of NTCP

Plain Language Summary

- Sodium taurocholate cotransporting polypeptide (NTCP) is a protein exclusively found on the surface of hepatocytes, or liver cells
- This protein is the major host receptor that allows hepatitis delta virus (HDV) to enter a hepatocyte, making it an important target for the treatment of HDV
- Bulevirtide blocks the entry of the HDV by binding to NTCP on the surface of hepatocytes
- Six NTCP single nucleotide polymorphisms were identified in 1 or more racial groups from public datasets
- In this study, NTCP expression levels and variants in 50 US primary human hepatocyte (PHH) donors were investigated. Our data demonstrated that bulevirtide remained active against HDV infection on the PHHs with different NTCP levels and NTCP variants

References: **1.** Yan H, et al. *Dig Dis.* 2015;33(3):388-96. **2.** Ni Y, et al. *Gastroenterology*. 2014;146:1070-83. **3.** Urban S, et al. *Gastroenterology*. 2014;147:48-64. **4.** Hepcludex. European Medicines Agency SmPC. Gilead Sciences, Inc.; 2023. **5.** Byrska-Bishop M, et al. *Cell.* 2022;185(18):3426-40. **6.** Chen S, et al. *bioRxiv*. 2022. https://doi.org/10.1101/2022.03.20.485034.

Acknowledgments: This study was funded by Gilead Sciences, Inc. Medical writing and editorial support were provided by Breanne E. Pirino, PhD, of AlphaScientia, a Red Nucleus company, and was funded by Gilead Sciences, Inc.

Disclosures: **SX**, **TA**, **SM**, **CL**, **RosM**, **YL**, **EM**, **HM**, and **RobM** are employees of Gilead Sciences, Inc., and may own stock in Gilead Sciences, Inc.

Introduction

Sodium Taurocholate Cotransporting Polypeptide

Figure 1. Sodium Taurocholate Cotransporting Polypeptide Structure

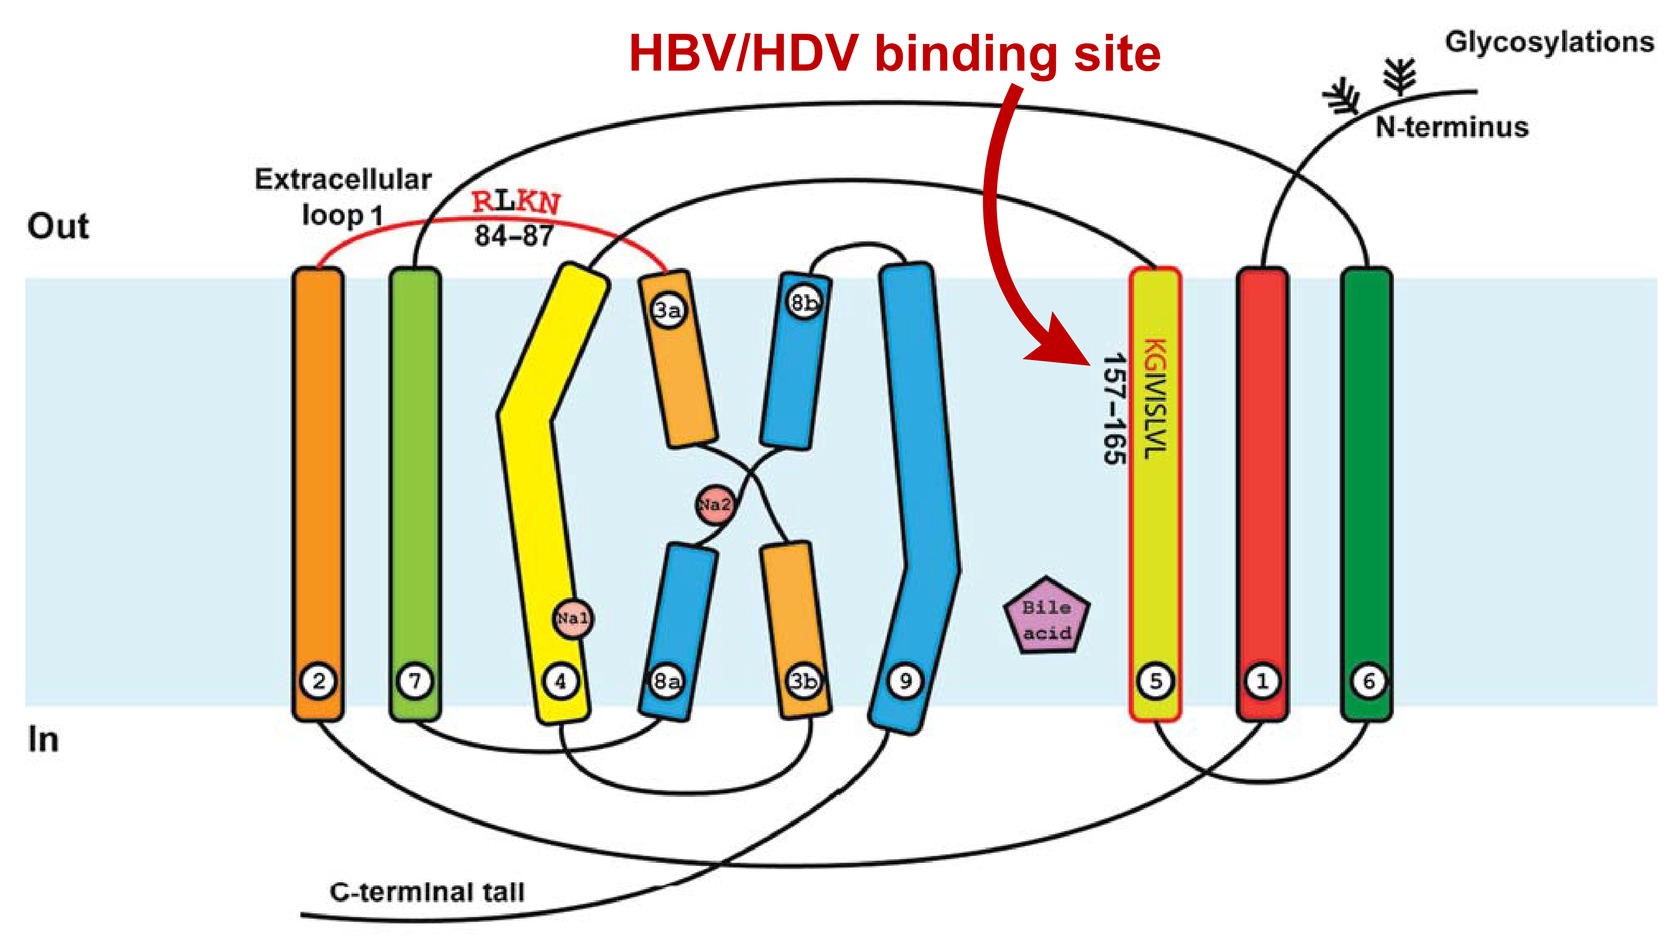

HBV/HDV binding site
Glycosylations
N-terminus
Extracellular loop 1
RLKN
84–87
Out
In
KGIVISLVL
157–165
Bile acid
C-terminal tail

HBV, hepatitis B virus; HDV, hepatitis delta virus.

- Sodium taurocholate cotransporting polypeptide (NTCP) is a 349–amino acid multipass transmembrane protein[1] with 5 exons
- Exclusively expressed at the membrane of hepatocytes
- Important for uptake of bile salts and selected drugs
- NTCP is the major host receptor for hepatitis B virus (HBV)/hepatitis delta virus (HDV) entry (**Figure 1**)[2]

Bulevirtide

Figure 2. BLV Blocks HDV Viral Entry

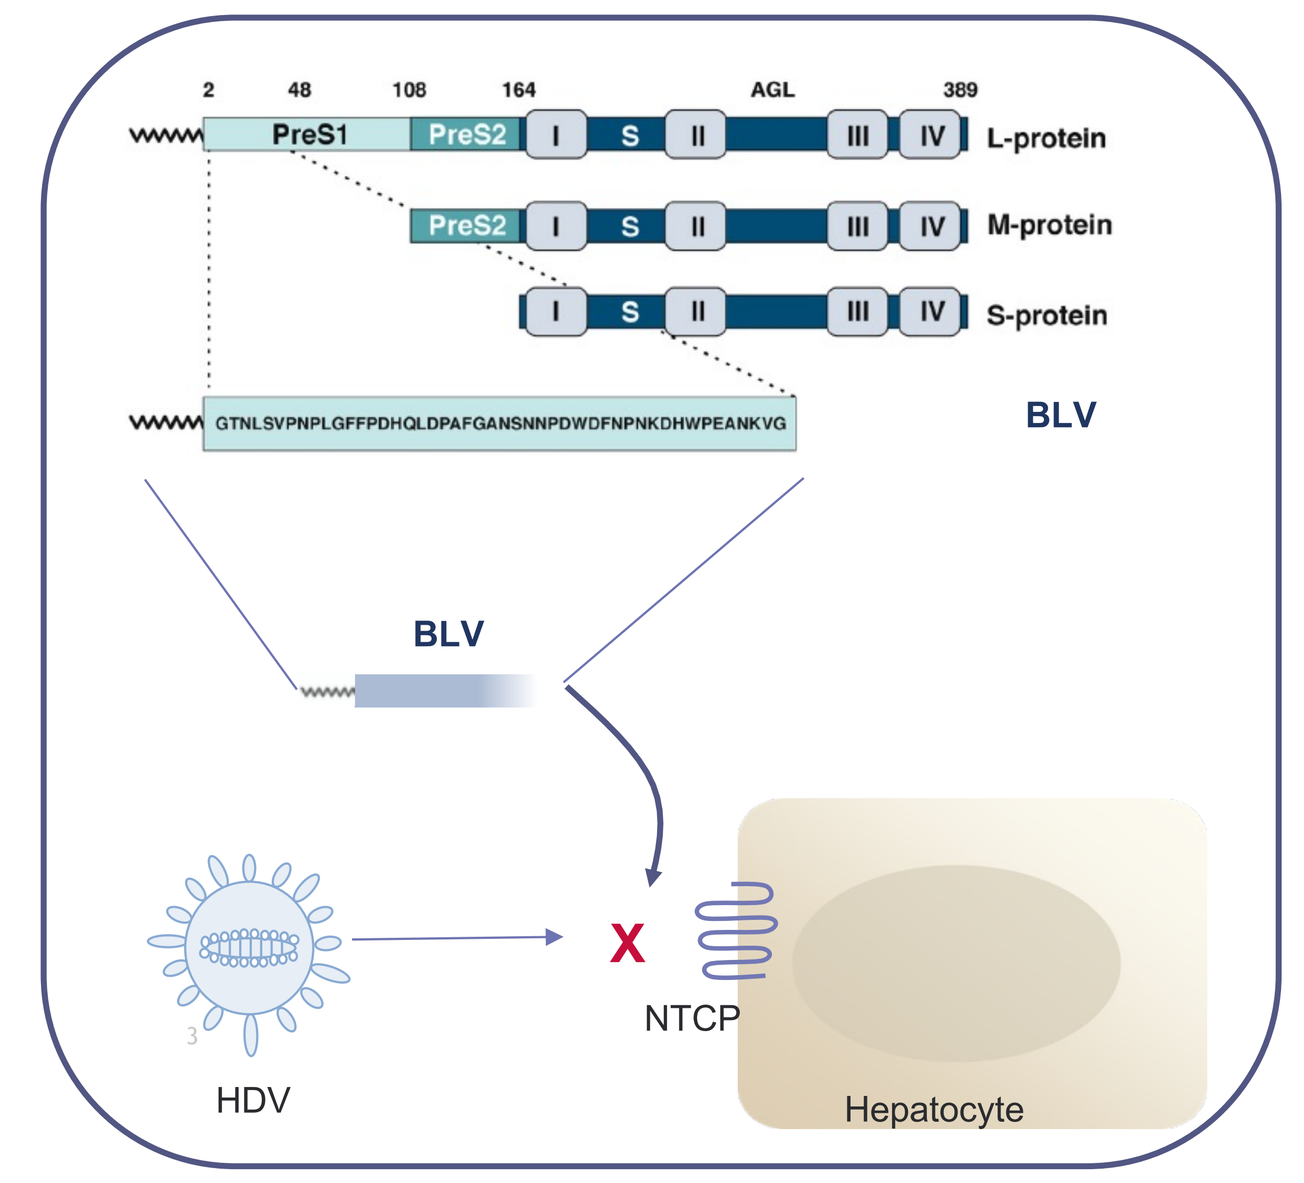

PreS1
PreS2
L-protein
M-protein
S-protein
AGL
BLV
GTNLSVPNPLGFFPDHQLDPAFGANSNNPDWDFNPNKDHWPEANKVG
X
HDV
NTCP
Hepatocyte

AGL, antigenic loop; BLV, bulevirtide; HDV, hepatitis delta virus; NTCP, sodium taurocholate cotransporting polypeptide.

- Bulevirtide (BLV) is composed of the 47 amino acids of the preS1 domain of the HBV envelope, and it directly binds to NTCP to block HDV from entering hepatocytes (**Figure 2**)[3]
- Fully approved as treatment for HDV in Europe[4]

Objective

- To investigate the roles of NTCP expression level and single nucleotide polymorphisms (SNPs) on BLV-mediated inhibition of HDV
 - To identify NTCP SNPs in public datasets
 - To investigate NTCP expression variations and SNPs for 50 primary human hepatocyte (PHH) donors representative of US racial groups and BLV susceptibility
 - To evaluate BLV susceptibility on Huh7-NTCP cell lines expressing different NTCP levels and the NTCP variant T503C (I168T)

Methods

Identification of NTCP SNPs in US Racial Groups

- NTCP SNPs were identified for the most common US racial groups in 2 datasets: the US dataset from the 1000 Genomes Project[5] and the global dataset gnomAD v.3.1.2[6]
- Minor allele frequencies (MAFs) were calculated for all SNPs in the coding region of NTCP by dividing the number of alleles with a variant by the total number of alleles
 - SNPs with a MAF >1% in at least 1 of the racial groups (White, Black, Asian, and Latino) in gnomAD v3.1.2 (noncancer) or in 1 of the selected 1000 Genomes populations were compiled

Determination of SNPs and NTCP Expression Levels for PHH Donors

- Sample: PHHs from 50 US donors
- NTCP polymorphism analysis: whole-exon sequencing
- NTCP protein expression quantification: fluorescence-activated cell sorting (FACS) and mean fluorescence intensity (MFI)

Generation of Cell Lines Expressing Different Levels of NTCP and NTCP Variant I168T

- Huh7-NTCP cell lines expressing different levels of NTCP were generated by transfection of a plasmid carrying wild-type NTCP to Huh7 cells
- The Huh7-NTCP-I168T cell line was generated by transfection of a plasmid carrying NTCP-I168T to Huh7 cells
- NTCP expression was assessed by FACS analysis

Phenotyping Assay of PHHs (Figure 3)

- On day 0, PHHs were seeded in a 96-well plate
- On day 1, PHHs were pretreated with BLV for 1 hour before HDV infection
- On day 6, HDV-infected PHHs were fixed
- Anti–hepatitis delta antigen (HDAg) immunofluorescent (IF) staining was performed, and BLV susceptibility was determined by quantification of percent of average intensity

Figure 3. In Vitro Phenotyping Assay

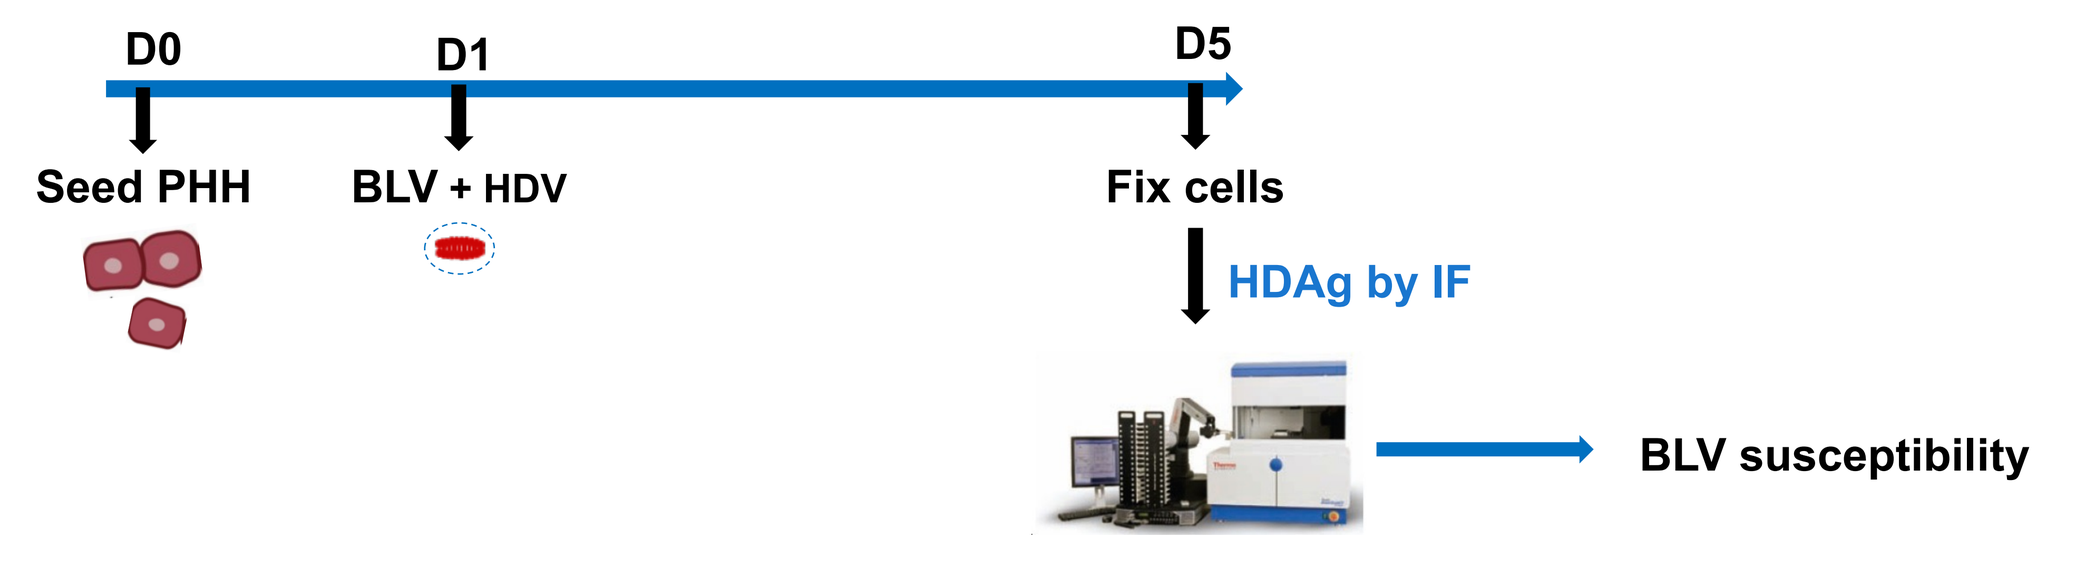

D0
D1
D5
Seed PHH
BLV + HDV
Fix cells
HDAg by IF
BLV susceptibility

BLV, bulevirtide; D, day; HDAg, hepatitis delta antigen; HDV, hepatitis delta virus; IF, immunofluorescence; PHH, primary human hepatocyte.

Results

Table 1. NTCP Polymorphism Prevalence From Public Dataset

NTCP SNPs		US NTCP Polymorphisms by Race[5]				Global NTCP Polymorphisms by Race[6]			
Variant	AA change	White n = 198	Black n = 122	Asian n = 206	Latino n = 128	White n = 64,812	Black n = 41,130	Asian n = 9940	Latino n = 15,100
800C>T	S267F	0.00%	0.00%	0.00%	0.00%	0.00%	0.10%	3.60%	0.10%
668T>C	I223T	0.50%	1.60%	0.00%	0.00%	0.01%	4.10%	0.01%	0.30%
225G>A	No change	10.60%	1.60%	9.20%	16.40%	9.00%	2.20%	14.00%	12.10%
627T>C	No change	0.00%	4.90%	0.00%	0.00%	0.07%	6.10%	0.00%	0.80%
111C>T	No change	0.00%	0.00%	1.90%	0.00%	0.00%	0.01%	0.80%	0.00%
960C>A	No change	0.00%	0.80%	0.00%	0.00%	0.01%	1.30%	0.00%	0.06%

AA, amino acid; NTCP, sodium taurocholate cotransporting polypeptide; SNP, single nucleotide polymorphism.

- Six NTCP SNPs (2 nonsynonymous and 4 synonymous) were observed in the public dataset with MAFs >1% in 1 or more racial groups (**Table 1**)

Table 2. Four of 6 NTCP SNPs From Public Dataset Identified in 50 PHH Donors

NTCP SNP	Amino Acid Change	Number of PHHs Carrying Variant	Race	BLV Binding Domain (157–165)
C800T	S267F	1/50	Asian	No
T668C	I223T	1/50	Black	No
G225A	T75T (no change)	4/50	White, Latino, Black	No
T627C	A209A (no change)	2/50	Black	No

BLV, bulevirtide; NTCP, sodium taurocholate cotransporting polypeptide; PHH, primary human hepatocyte; SNP, single nucleotide polymorphism.

- Two of 6 NTCP SNPs (111C>T and 960C>A) from public datasets were not identified in the 50 PHH donors (**Table 2**)
- These 2 SNPs did not result in amino acid changes and had a low prevalence (<2%) in all racial groups
- The SNPs were not tested for BLV susceptibility

Figure 4. NTCP Protein Levels Varied 5.8-Fold Among 50 PHH Samples From US Donors

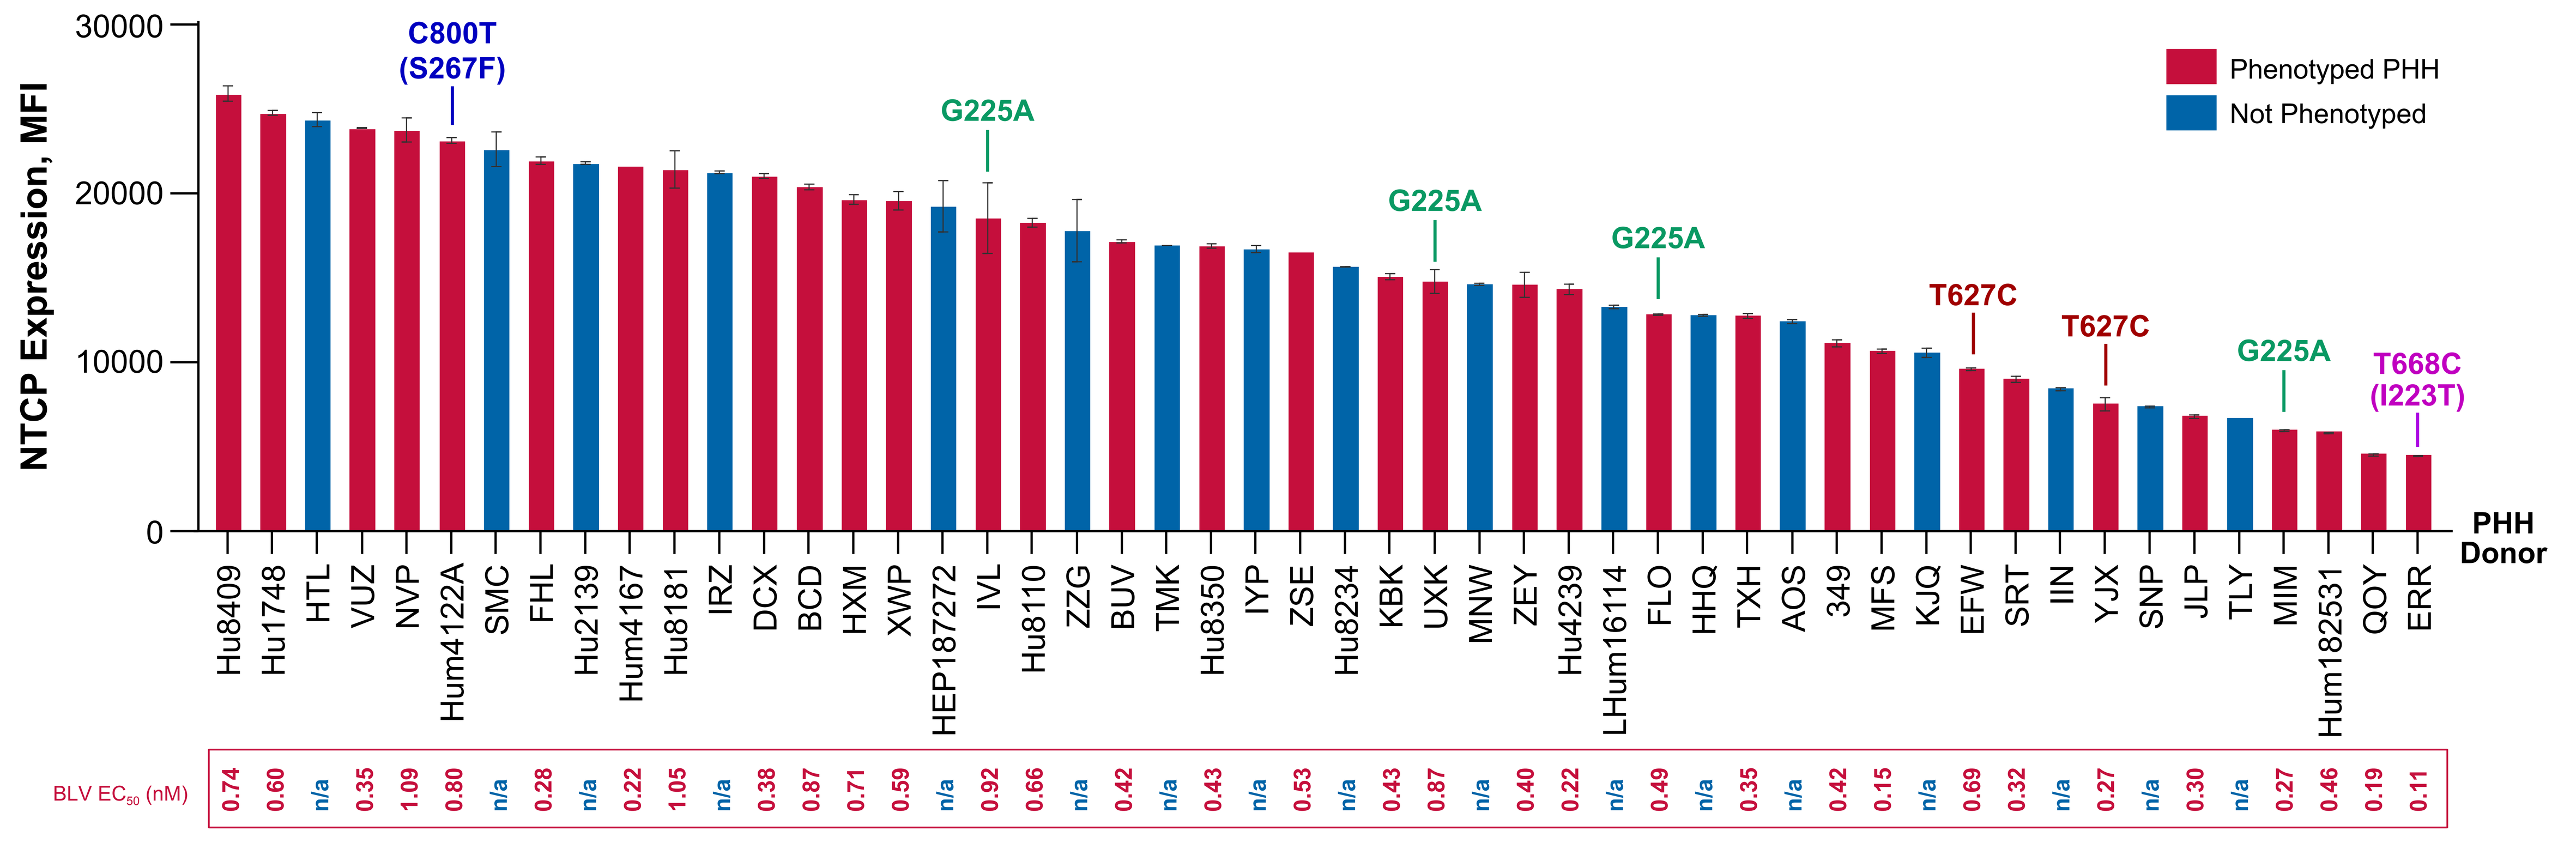

NTCP Expression, MFI
30000
20000
10000
0
C800T (S267F)
G225A
G225A
G225A
T627C
T627C
G225A
T668C (I223T)
Phenotyped PHH
Not Phenotyped
PHH Donor
Hu8409 Hu1748 HTL VUZ NVP Hum4122A SMC FHL Hu2139 Hum4167 Hu8181 IRZ DCX BCD HXM XWP HEP187272 IVL Hu8110 ZZG BUV TMK Hu8350 IYP ZSE Hu8234 KBK UXK MNW ZEY Hu4239 LHum16114 FLO HHQ TXH AOS 349 MFS KJQ EFW SRT IIN YJX SNP JLP TLY MIM Hum182531 QOY ERR
BLV EC50 (nM)
0.74 0.60 n/a 0.35 1.09 0.80 n/a 0.28 n/a 0.22 1.05 n/a 0.38 0.87 0.71 0.59 n/a 0.92 0.66 n/a 0.42 n/a 0.43 n/a 0.53 n/a 0.43 0.87 n/a 0.40 0.22 n/a 0.49 n/a 0.35 n/a 0.42 0.15 n/a 0.69 0.32 n/a 0.27 n/a 0.30 n/a 0.27 0.46 0.19 0.11

BLV, bulevirtide; EC_{50}, half-maximal effective concentration; MFI, mean fluorescence intensity; n/a, not available; NTCP, sodium taurocholate cotransporting polypeptide; PHH, primary human hepatocyte.

- NTCP protein expression was evaluated by FACS analysis and quantified with MFI
- MFI of the PHHs across the 50 donors ranged from 4447 to 25,934 (**Figure 4**)
- Except for donor ERR with T668C (I223T), the rest of the PHHs with the NTCP variants had MFIs ranging from 5964 to 23,155, suggesting NTCP variants have no major effect on NTCP protein expression
- Half-maximal effective concentrations (EC_{50}) of BLV were determined on PHHs with the entire range of NTCP expression and the 4 NTCP SNPs

Table 3. NTCP Protein Expression and BLV EC_{50} of Different Cell Lines

Cell Line	Characteristic of Cell Line	NTCP Protein Expression (MFI)	BLV EC_{50} (nM)
Human fibroblast	No NTCP receptor (background control)	125	n/a
Huh7	Parental cell line with extremely low level of NTCP	206	n/a
Huh7_NTCP_L	Cell line expressing low level of NTCP	2654	0.40
Huh7_NTCP_H	Cell line expressing high level of NTCP	14,716	0.98
Huh7-NTCP-I168T	Cell line expressing NTCP-I168T	3289	0.20

n/a, BLV EC_{50} is not available due to no infection by HDV.
BLV, bulevirtide; EC_{50}, half-maximal effective concentration; HDV, hepatitis delta virus; MFI, mean fluorescence intensity; NTCP, sodium taurocholate cotransporting polypeptide.

- Huh7 cells expressing different NTCP levels or NTCP-I168T remained sensitive against HDV (**Table 3**)

Figure 5. Sensitivity of BLV Against HDV on PHHs With Different NTCP Levels and Variants

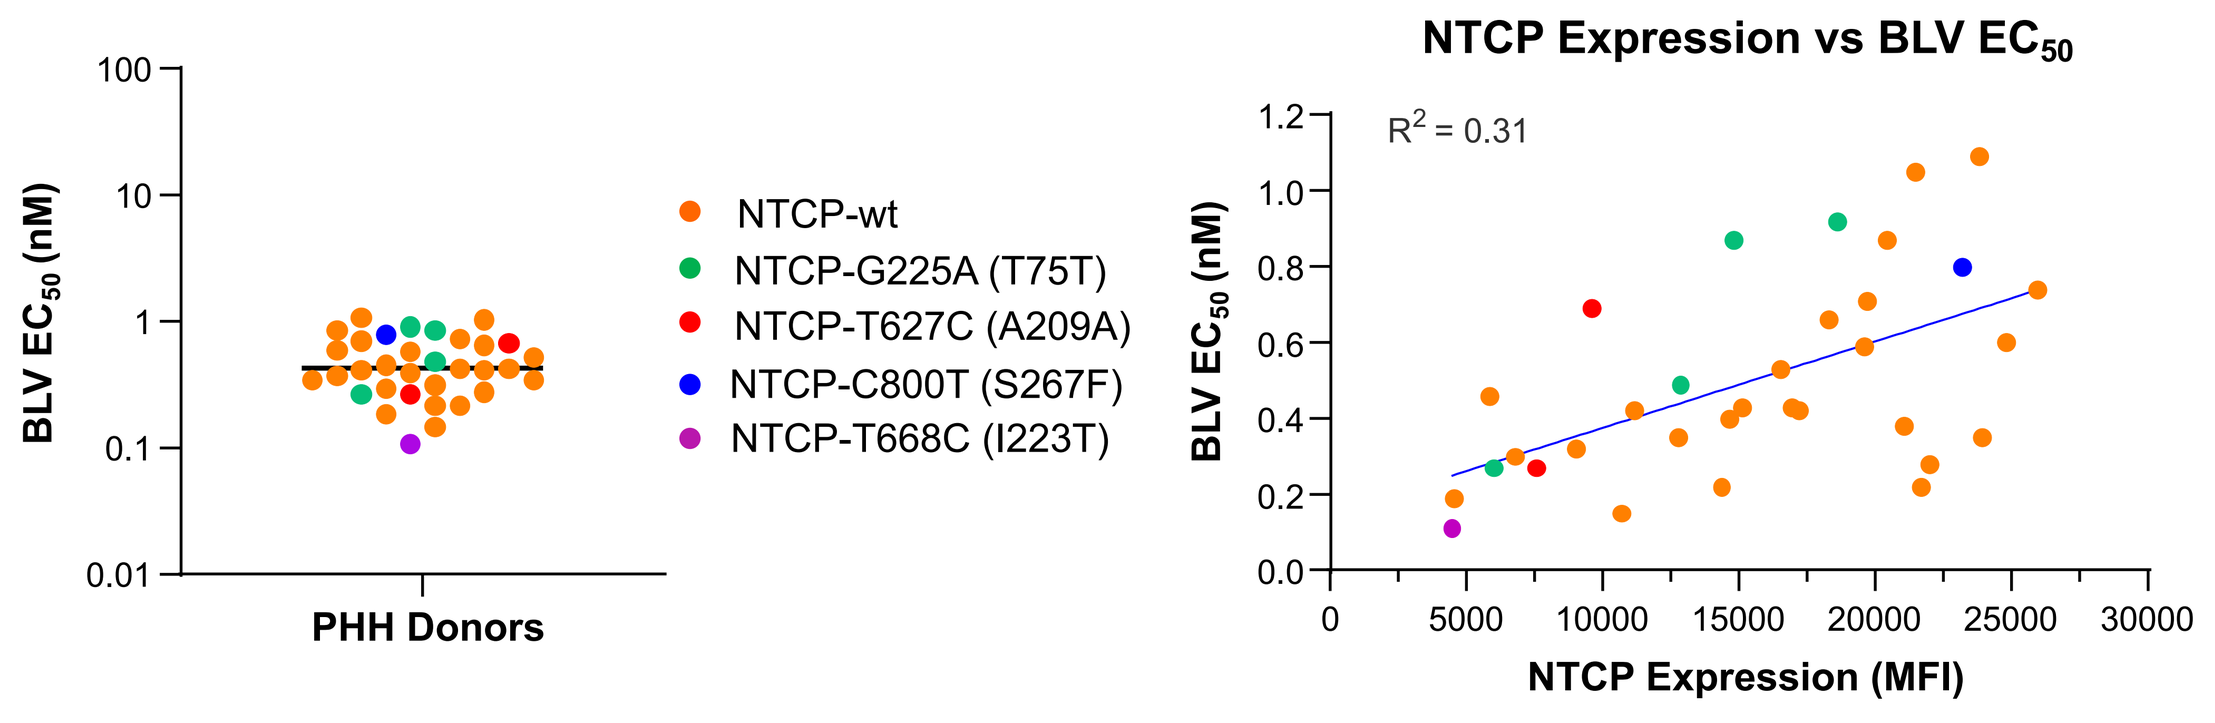

BLV EC50 (nM)
100
10
1
0.1
0.01
PHH Donors
NTCP-wt
NTCP-G225A (T75T)
NTCP-T627C (A209A)
NTCP-C800T (S267F)
NTCP-T668C (I223T)
NTCP Expression vs BLV EC50
R2 = 0.31
1.2
1.0
0.8
0.6
0.4
0.2
0.0
0 5000 10000 15000 20000 25000 30000
NTCP Expression (MFI)

BLV, bulevirtide; EC_{50}, half-maximal effective concentration; HDV, hepatitis delta virus; MFI, mean fluorescence intensity; NTCP, sodium taurocholate cotransporting polypeptide; PHH, primary human hepatocyte; wt, wild-type.

- BLV remained sensitive against HDV on PHHs expressing different levels of NTCP and NTCP variants (**Figure 5**)

European Association for the Study of the Liver; 05–08 June 2024; Milan, Italy